779TiP: Commencement of First-in-human Phase 1/2 TCR-T Clinical Trial Targeting Shared Tumor-specific Hotspot Mutations in Solid Tumors

Maria Pia Morelli[1], Marcelo V. Negrao[1], Amir A. Jazaeri[1], Benny Johnson[1], Partow Kebriaei[1], Drew C. Deniger[2], Matthew R. Collinson-Pautz[2], Nathan A. Demars[2], John V. Heymach[1], Scott Kopetz[1]
[1]MD Anderson Cancer Center, Houston, TX, USA; [2]Alaunos Therapeutics, Inc. Houston, TX, USA

Background

Adoptive T-cell therapy is an emerging strategy for solid tumors. Cancer cells frequently harbor driver-mutations in *KRAS, TP53,* and *EGFR* genes that can be targeted by T-cell receptors (TCRs). These neoepitopes are presented on the tumor cell surface by human leukocyte antigen (HLA) molecules to TCRs. We have developed, using non-viral *Sleeping Beauty* transposition, a library of TCRs able to target *KRAS*, *TP53* and *EGFR* mutations for the treatment of solid tumors.

Neoantigen-Specific TCR-T Development

The *Sleeping Beauty* transposase/transposon system can be used as a non-viral gene transfer system in human cells. *Sleeping Beauty* transposase is briefly expressed to integrate the transposon into the genome and is then degraded and eliminated from the T cell. *Sleeping Beauty* transposon is inserted into TA dinucleotide repeats randomly within the human genome (**Figure 1A**). Co-transfer of *Sleeping Beauty* transposase and transposon into the T cell results in rapid and stable expression of the introduced neoantigen-specific TCR, which allows tumor cell recognition (**Figure 1B**). The *Sleeping Beauty system* has high flexibility, and low manufacturing time and cost compared to other gene transfer technologies.

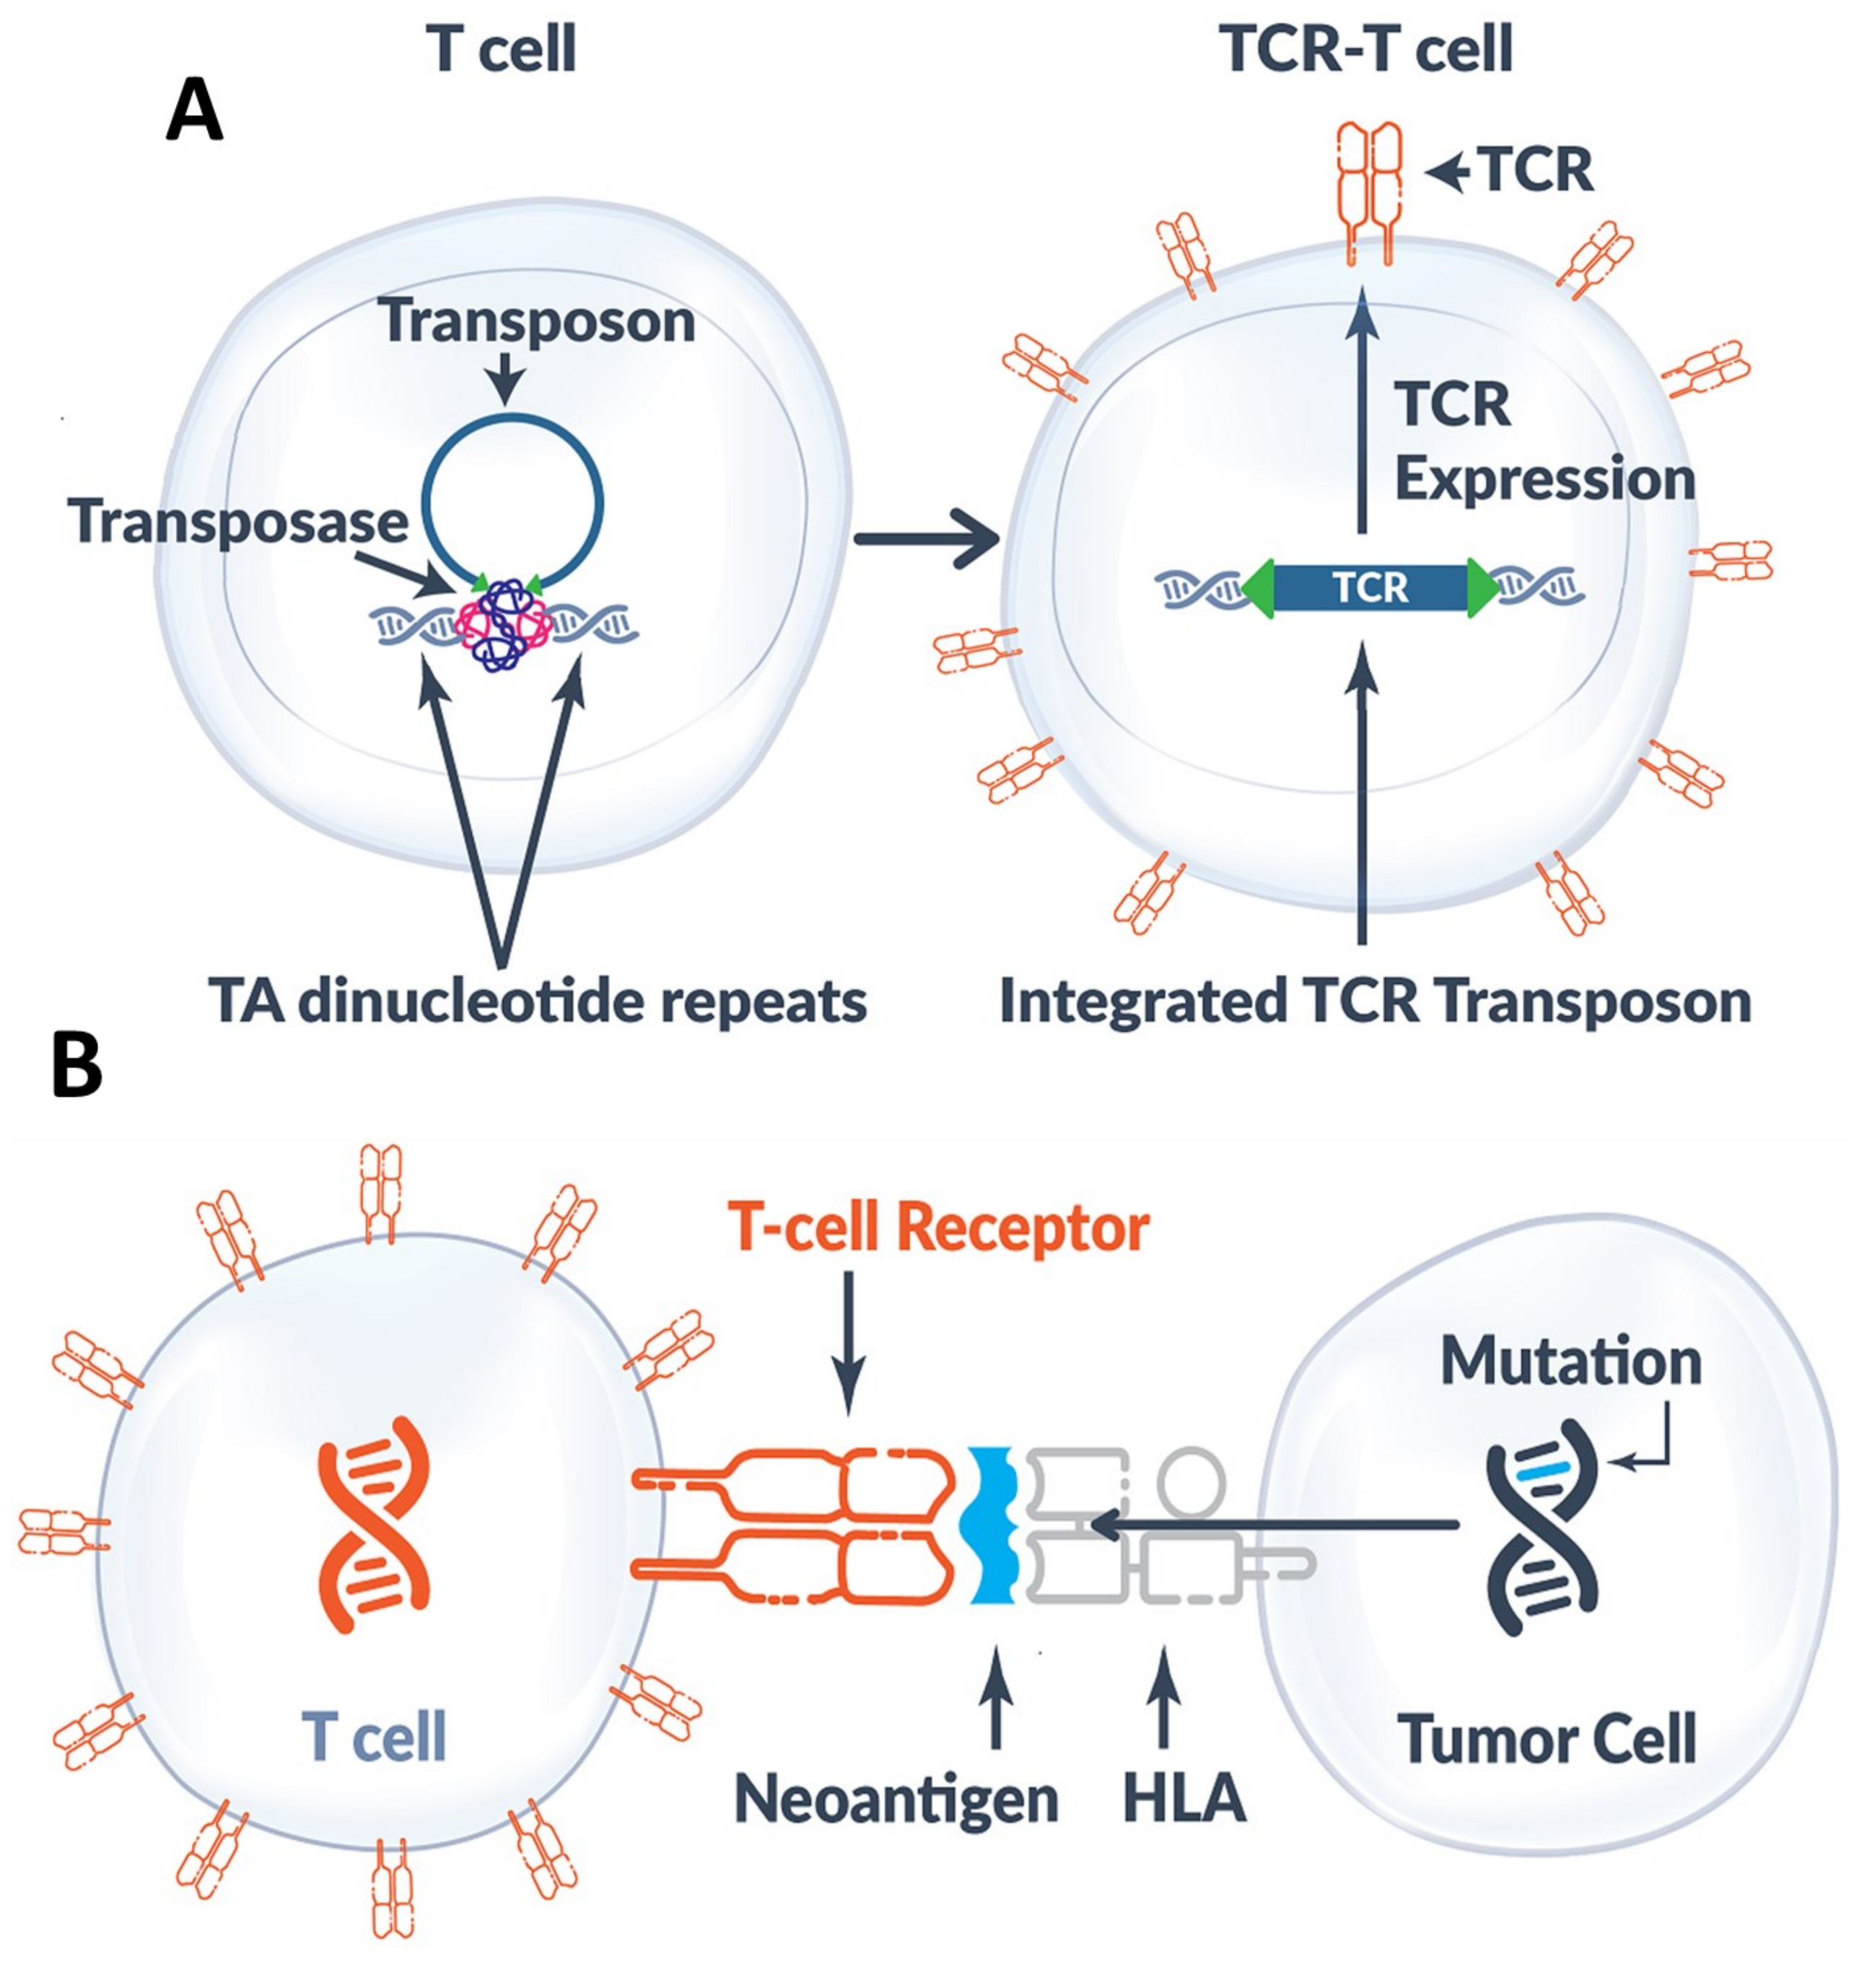
A
T cell
TCR-T cell
TCR
Transposon
Transposase
TCR
Expression
TCR
TA dinucleotide repeats
Integrated TCR Transposon
B
T-cell Receptor
Mutation
T cell
Neoantigen
HLA
Tumor Cell

Figure 1. A) Transposon / transposase system for integration into T cell DNA; **B)** tumor neoantigen recognition by transposed T cell receptor.

Alaunos Therapeutic's TCR Library - Figure 2

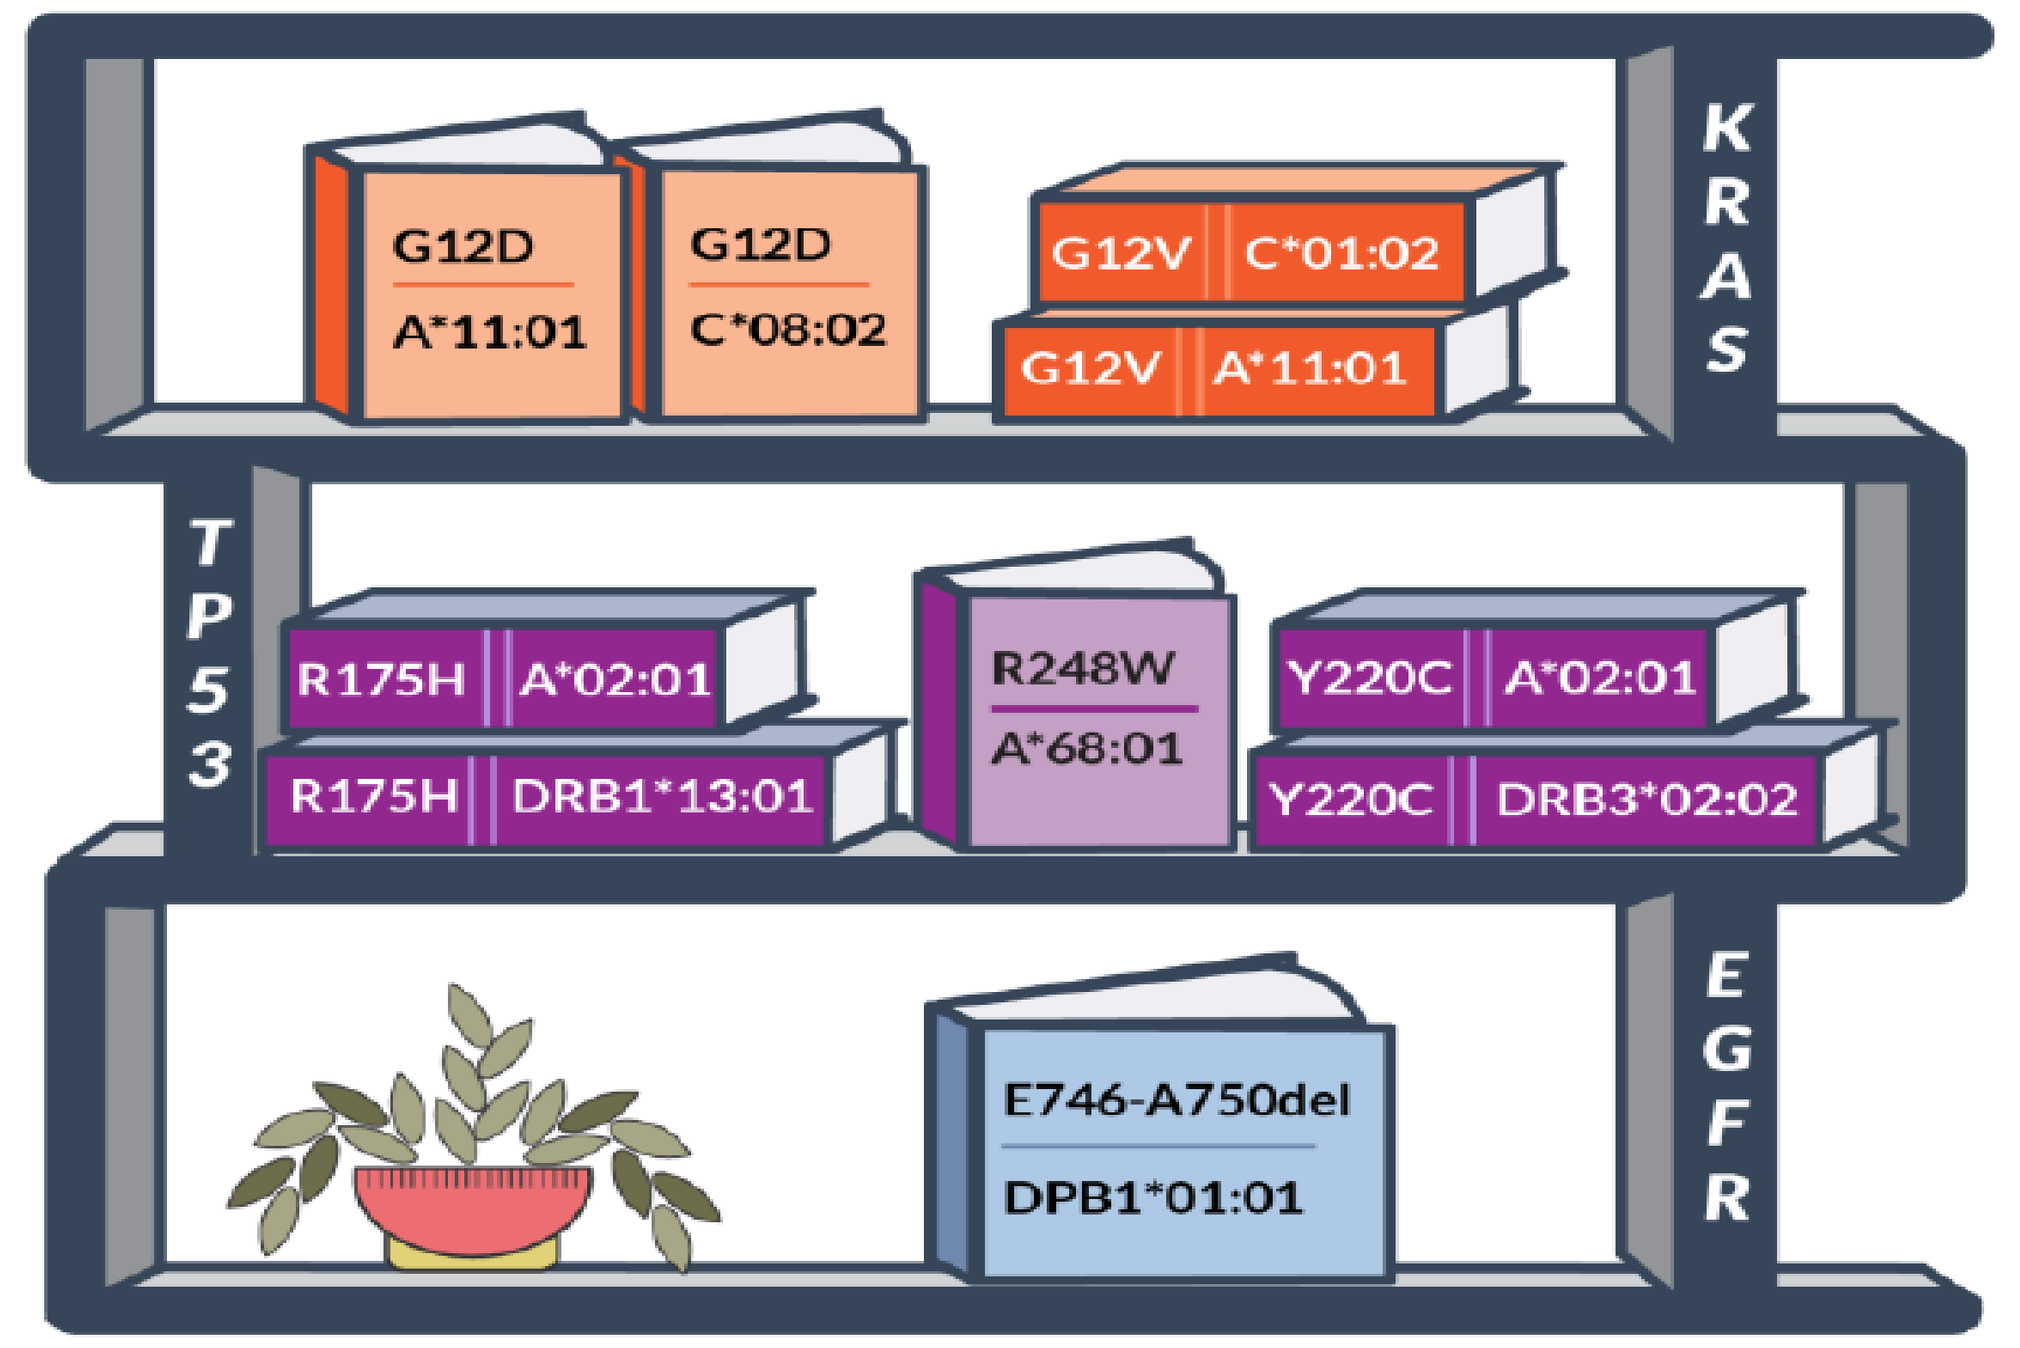
KRAS
G12D A*11:01
G12D C*08:02
G12V C*01:02
G12V A*11:01
TP53
R175H A*02:01
R175H DRB1*13:01
R248W A*68:01
Y220C A*02:01
Y220C DRB3*02:02
EGFR
E746-A750del DPB1*01:01

Study Design

This is a first in human phase 1/2 study of TCR-T cell therapy for patients with non-small cell lung, colorectal, endometrial, pancreatic, ovarian and bile duct cancer. Eligible patients have received and failed standard of care therapy for their tumor and have a mutation and HLA type match for a TCR in the library (**Figure 2**). After enrollment, patients will undergo leukapheresis, optional bridging therapy during manufacturing (**Figure 3**) and lymphodepletion. The patient's TCR-T cells will be administered to the patient intravenously following lymphodepletion. The starting dose will be 5 x 10^9 TCR-T cells (Dose Level 1 (DL1)), in the absence of a Dose Limiting Toxicity (DLT) escalating to 4 x 10^{10} (DL2) and 1 x 10^{11} (DL3) TCR-T cells, as safety allows. After completion of DL2, an analysis of TCR-T cell persistence will be performed, and, if deemed necessary, interleukin-2 (IL-2) will be administered after the TCR-T cell infusion. The primary objective of the phase I portion is to define the incidence of DLTs and the maximum tolerated dose (MTD) or recommended phase II dose (RP2D) of TCR-T cells administered without IL-2 (Arm A) or with IL-2 (Arm B). Clinical and radiologic responses will be assessed by RECIST (v1.1) at six and 12 weeks after TCR-T cell infusion and every 12 weeks thereafter for up to two years or until study discontinuation, whichever occurs first. All patients will continue to be monitored in a long-term follow-up protocol for up to 15 years post-TCR-T cell infusion (**Figure 4**). The study has been initiated and enrollment is ongoing.

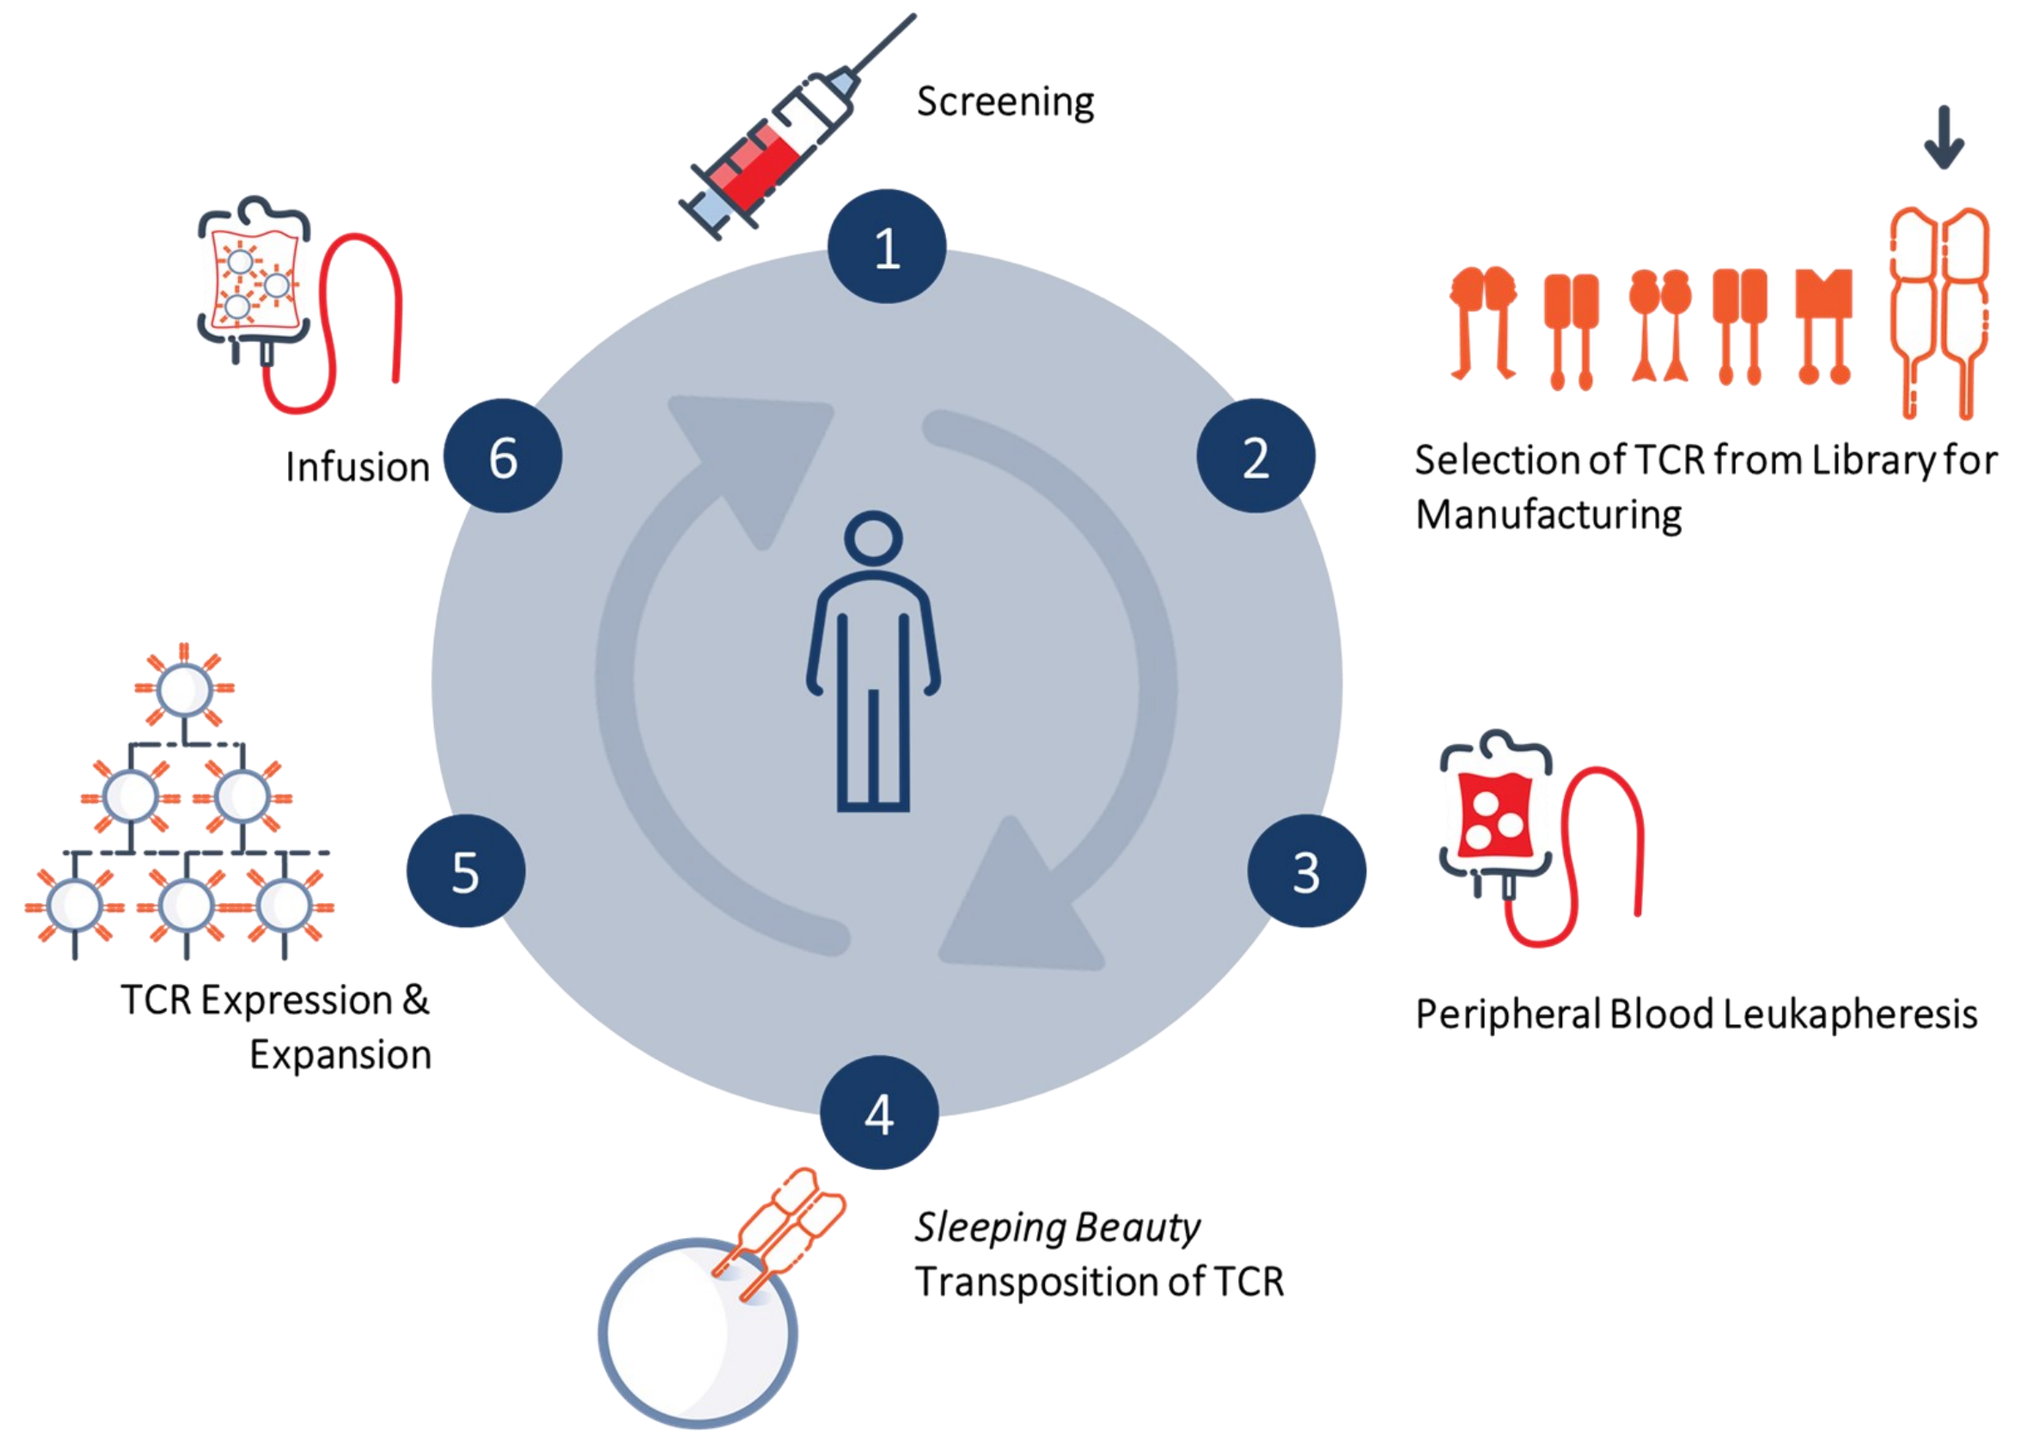
Screening
1
Selection of TCR from Library for Manufacturing
2
Peripheral Blood Leukapheresis
3
Sleeping Beauty Transposition of TCR
4
TCR Expression & Expansion
5
Infusion
6

Figure 3: Manufacturing process of autologous TCR-T cell drug product

Study Design - Figure 4

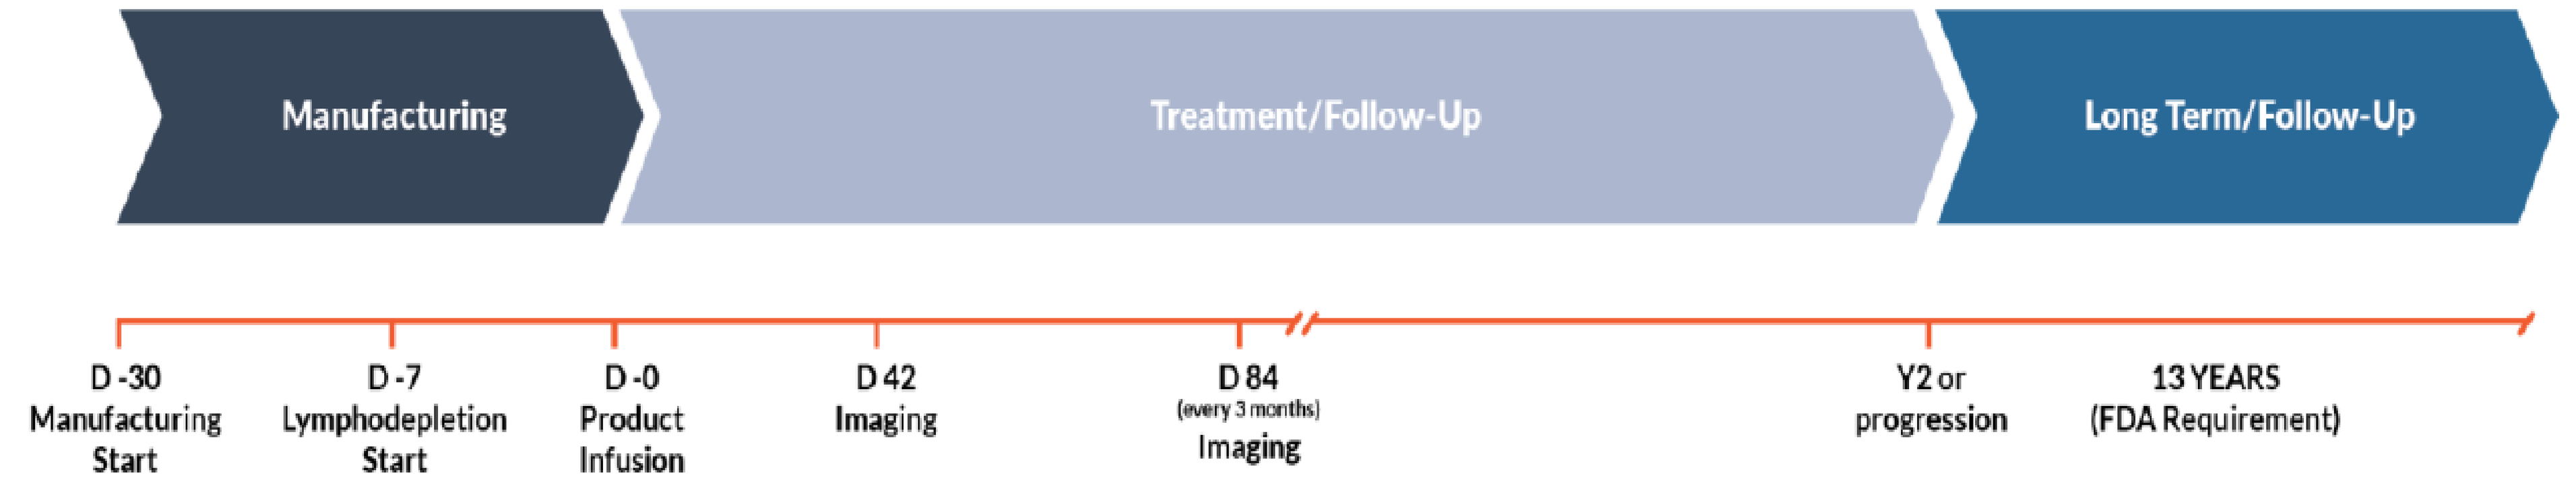
Manufacturing
Treatment/Follow-Up
Long Term/Follow-Up
D -30 Manufacturing Start
D -7 Lymphodepletion Start
D -0 Product Infusion
D 42 Imaging
D 84 (every 3 months) Imaging
Y2 or progression
13 YEARS (FDA Requirement)

Study Population and Key Inclusion / Exclusion Criterion

Key Inclusion Criteria

1. Patients with tumors that have somatic mutation(s) and HLA type restriction combination matching an available TCR in TCR library.
2. Patients who have previously received at least one line of standard systemic therapy for their advanced/metastatic cancer and have either progressed, recurred, or were intolerant to prior treatment.
 - Subgroup 1: Gynecologic cancers: a) Ovarian cancer: Subjects who are platinum-resistant, defined as progression on or within 6 months of prior platinum-based regimen; b) Endometrial cancer: Subjects who have received at least 2 prior lines of therapy for advanced/recurrent disease (includes adjuvant chemotherapy and/or chemo-radiation; prior hormonal therapy not included).
 - Subgroup 2: CRC: At least 2 prior lines of systemic treatment for advanced unresectable or metastatic disease, which must include an irinotecan or oxaliplatin-based therapy and, if eligible, a targeted antibody therapy. Subjects with deficient DNA mismatch repair or microsatellite instability-high CRC must have received treatment with an immune checkpoint inhibitor.
 - Subgroup 3: Pancreatic cancer: Subjects who have progressive disease after receiving one prior line of therapy (e.g., FOLFIRINOX, gemcitabine-based therapy).
 - Subgroup 4: NSCLC: Subjects with recurrent and/or metastatic disease with disease progression or intolerance to treatment with a PD-1/PD-L1 inhibitor either as a single-agent, or in combination with other immune checkpoint inhibitors (e.g., CTLA-4 inhibitors), and/or platinum-doublet chemotherapy. Subjects with targetable oncogene alterations (e.g., EGFR, ALK, ROS1, RET, MET, NTRK1-3, BRAF) must have had disease progression or intolerance to at least one prior line of targeted therapy.
 - Subgroup 5: Cholangiocarcinoma: Subjects must have histologically confirmed cholangiocarcinoma stage II, III, or IV (intra-hepatic, extra-hepatic and perihilar) that is not eligible for curative resection, transplantation, or ablative therapies, and who have progressed after receiving at least one line of standard therapy.
3. Patients must have evaluable or measurable disease per RECIST 1.1 with at least one lesion that can be measured that is not the biopsied lesion.

Key Exclusion Criteria

1. Known active CNS metastases
2. Concurrent systemic steroid therapy at a dose of >10 mg prednisone daily or equivalent is excluded.
3. Any form of primary immunodeficiency
4. Severe chronic respiratory condition.

Summary

- Applying the clinical *Sleeping Beauty* transposon/transposase **non-viral** system to generate neoantigen-specific TCR-T cells targeting non-small cell lung, colorectal, endometrial, pancreatic, ovarian, and bile duct cancer
- This proprietary **non-viral gene transfer** platform called *Sleeping Beauty* is utilized to genetically modify the patient's own T cells (both CD4+ and CD8+) using the TCR plasmid.
- The major **advantage** of using **non-viral vectors** is its potential bio-safety (e.g. minimizing insertional mutations/possible secondary malignancies)
- **Non-viral vectors** have also drawn significant attention due to their lower immunotoxicity.
- Phase I portion of the study is expected to complete accrual by the end of 2023
- Phase II portion will include expansion cohorts of non-small cell lung, colorectal, endometrial, pancreatic, ovarian, and bile duct cancer

- **Study Sponsored by: Alaunos Therapeutics Inc.**
- **Clinical Trial NCT Number: NCT05194735**
- **Contact information: Nate Demars, ndemars@alaunos.com**