hunTR™: a hyperplex platform for the discovery of neoantigen-reactive T-cell receptors

Guowei Gu[1], Julissa Simmons[1], Kristen Rimaila[1], Gaby Garland[1], Ugochi Ibekwe[1], Donghyun Joo[1], An Lu[1], Amanda Montoya[1], Pretty R. Mathew[1], Charles Ho[1], Hao Zhao[1],
Beatriz A. Santillan[1], Matthew R. Collinson-Pautz[1], & Drew C. Deniger[1,2]
[1] Alaunos Therapeutics, Inc., Houston TX [2] Corresponding Author

TCR-T cell therapy for targeting solid tumors

Solid tumors harbor mutations that can give rise to neoantigens recognized by T-cell receptors (TCRs) expressed on tumor infiltrating lymphocytes (TILs). Alaunos has developed a library of TCRs targeting hotspot mutations based on the non-viral *Sleeping Beauty* transposon/transposase system, which is presently being evaluated in a first-in-human phase 1/2 study in patients with non-small cell lung, colorectal, endometrial, pancreatic, ovarian, and bile duct cancers. Current methods of TCR discovery require large volume blood draws or immortalization of primary cells with live viruses to make antigen presenting cells (APCs), limiting the ability to efficiently discover new TCRs and is only applicable to some patients. Alaunos developed hunTR™ (human neoantigen T-cell Receptor), a rapid, hyperplex platform for the discovery of neoantigen-reactive TCRs from limited starting material.

hunTR™ is designed to identify neoantigen reactive TCRs

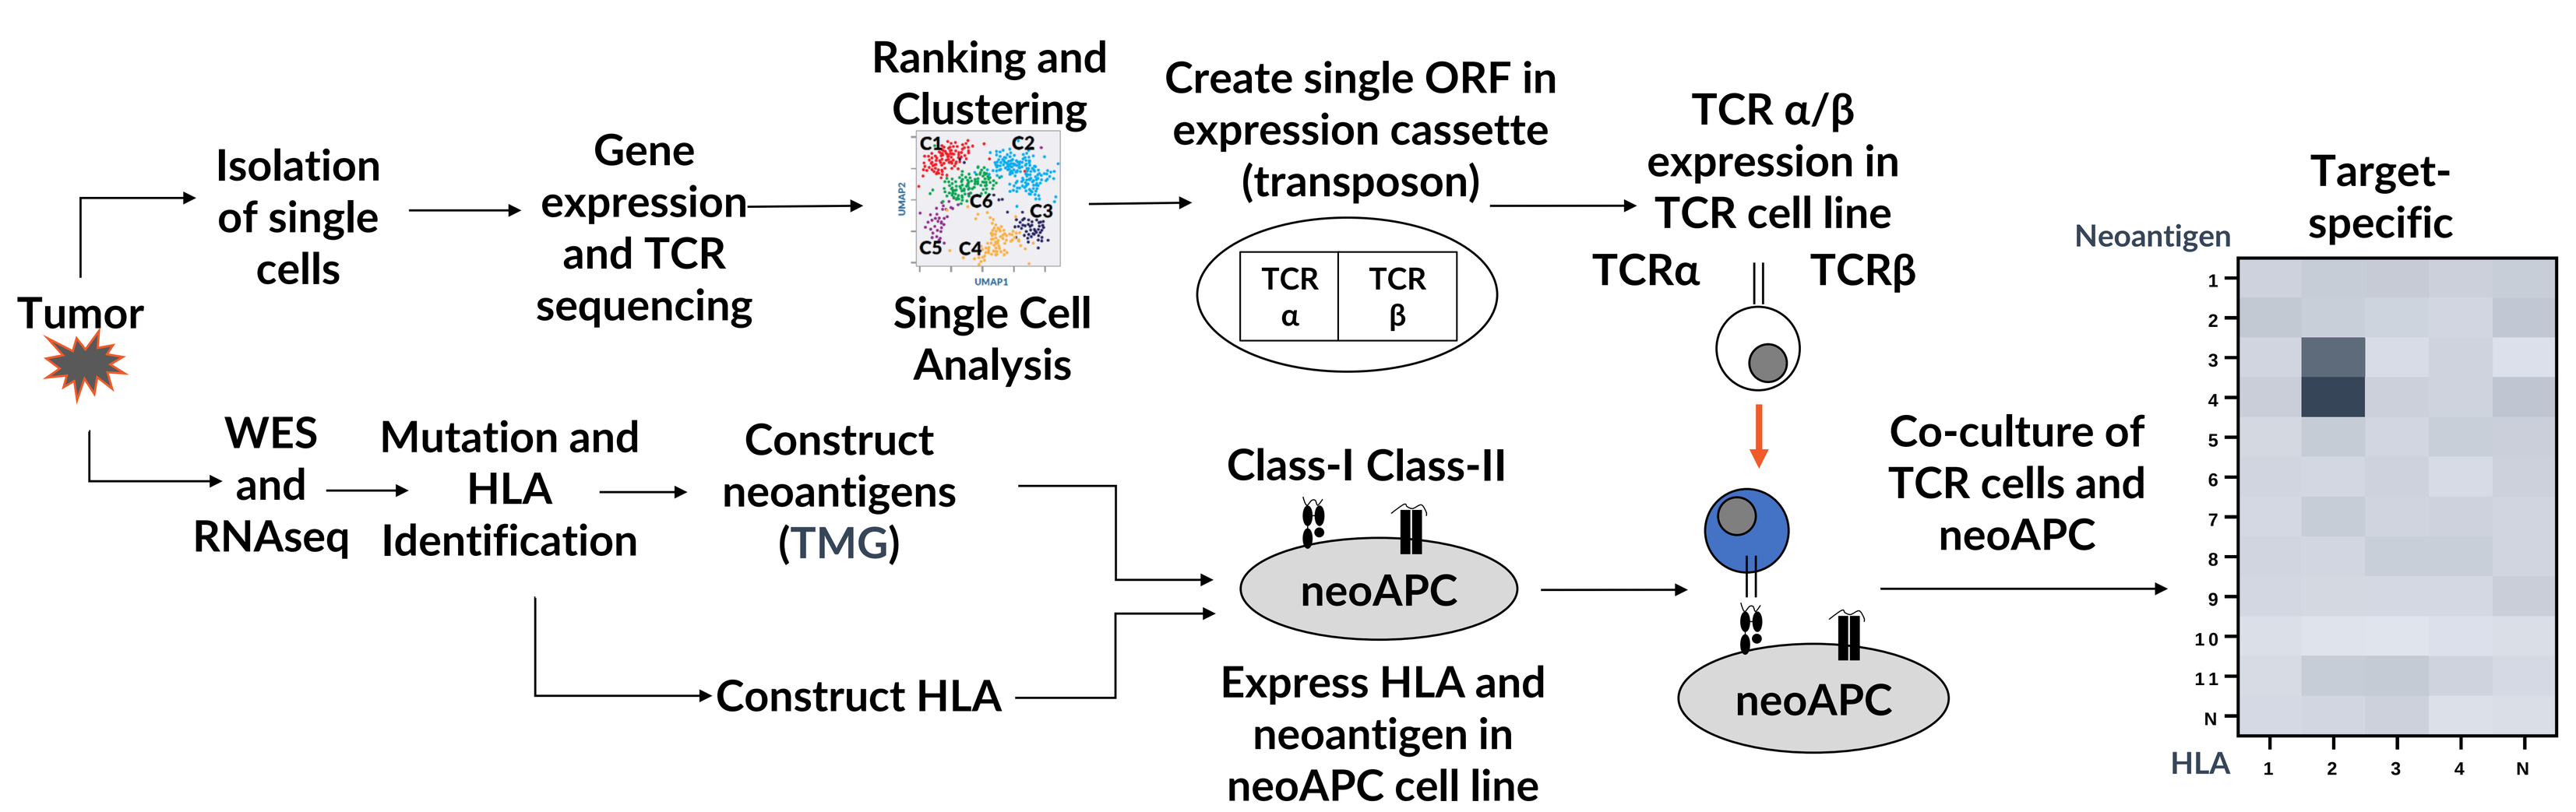
Tumor
Isolation of single cells
Gene expression and TCR sequencing
Ranking and Clustering
Single Cell Analysis
Create single ORF in expression cassette (transposon)
TCR α
TCR β
TCR α/β expression in TCR cell line
TCRα
TCRβ
WES and RNAseq
Mutation and HLA Identification
Construct neoantigens (TMG)
Construct HLA
Class-I Class-II
neoAPC
Express HLA and neoantigen in neoAPC cell line
neoAPC
Co-culture of TCR cells and neoAPC
Neoantigen
Target-specific
HLA

TILs sorted from digested tumor specimens were processed to obtain TCR sequences and gene expression profiles on a single cell basis. These data were fed into a novel bioinformatics pipeline to identify TCRs with predicted neoantigen reactivity. TCRs were reconstructed in *Sleeping Beauty* transposon plasmids and expressed in TCR cells, an engineered cell line capable of detecting TCR reactivity to both class I and class II HLA-restricted neoepitopes. Somatic single nucleotide variants, short insertions/deletions, and germline class I and II HLA alleles were called for each patient. neoAPCs were engineered to express relevant patient-derived neoantigens and HLA molecules. TCR cells were cocultured with a matrix of neoAPCs, and conditions yielding a neoantigen reactivity were identified.

Single cell RNA seq clustering informs TCR selection for screening

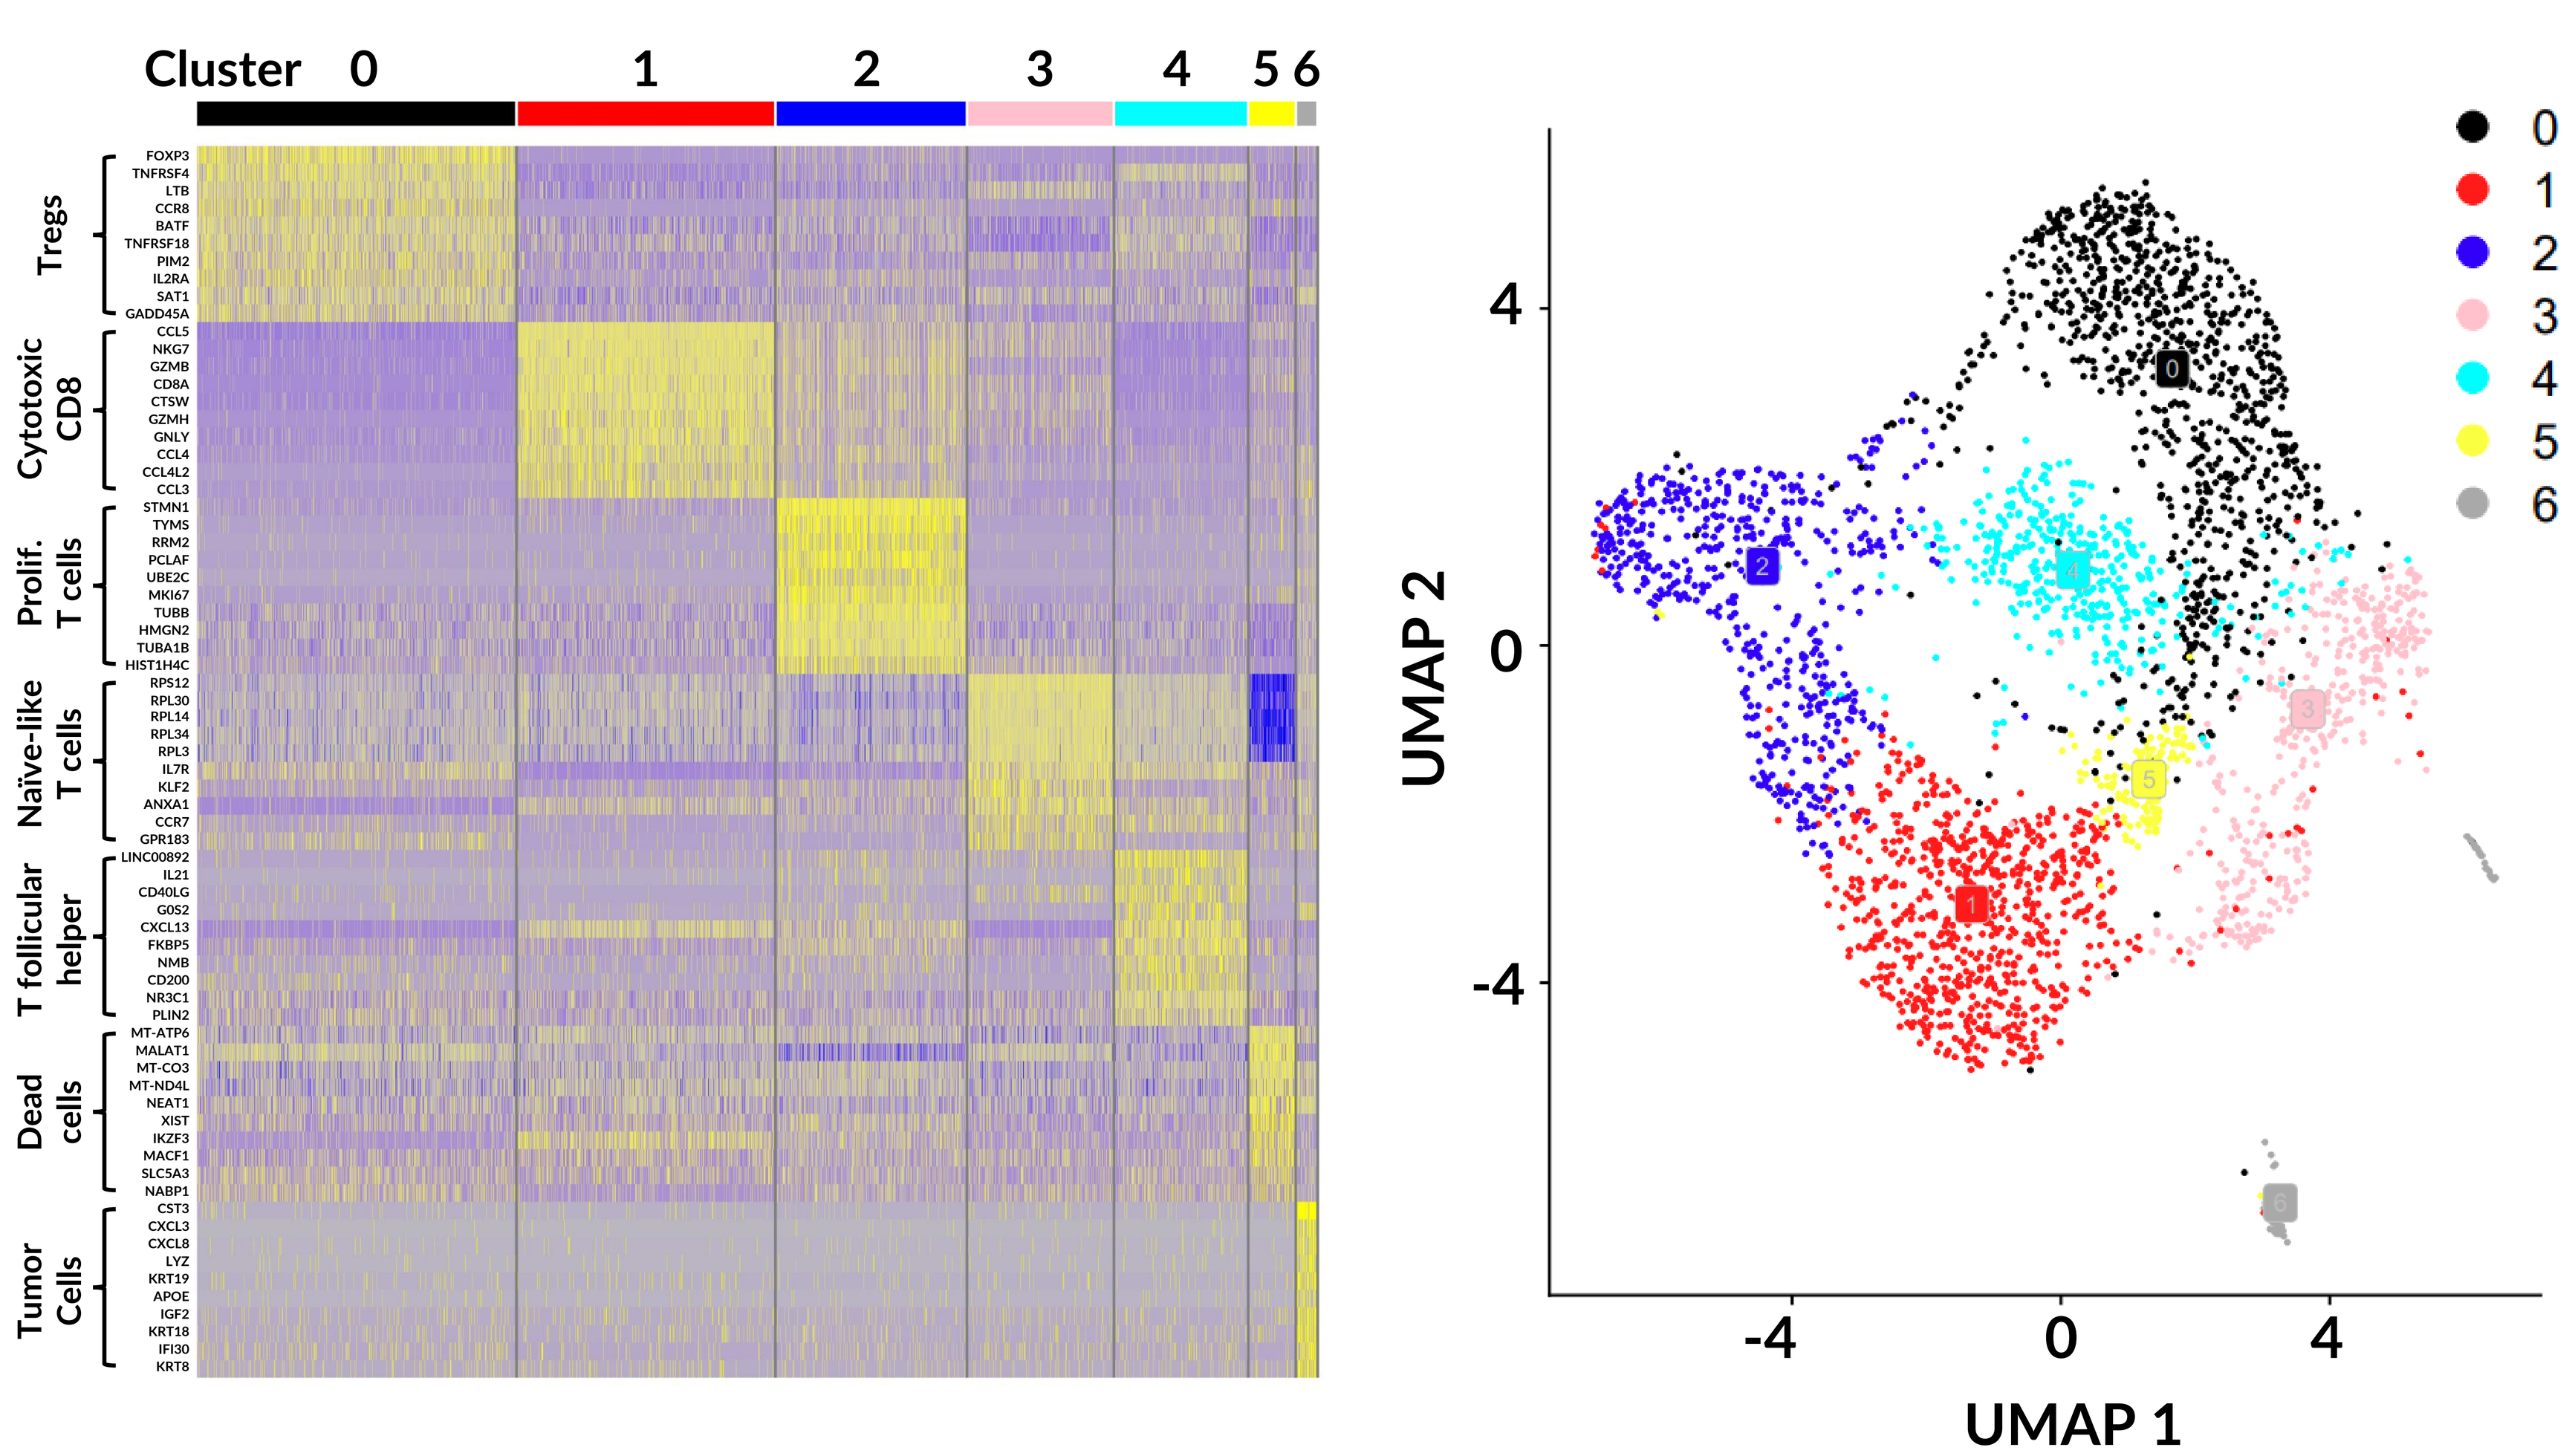
Cluster 0 1 2 3 4 5 6
Tregs
Cytotoxic CD8
Prolif. T cells
Naive-like T cells
T follicular helper
Dead cells
Tumor Cells
UMAP 2
UMAP 1

Single cell gene expression and TCR sequencing data of TIL were quality controlled and processed through bioinformatic pipelines and dimensionality reduction and clustering methods to identify TCR candidates for screening.

Mutation reactive TCRs were found in all 9 patients screened across 3 tumor types

Patient ID	Cancer Diagnosis	# Mutations Screened	# HLA Screened	# TCRs Screened	Total Plexity of Screening	TCR Detection Rate % (N)	Reactive TCR HLA Restriction	Neoantigen Recognized by TCR
2599	Colorectal	75	15	18	20,250	5.6% (1)	Class I (HLA-A)	ERGIC2 L176P
6932	Breast	33	16	22	11,616	4.5% (1)	Class II (HLA-DP)	HELZ2 p.P775A
5239	Colorectal	101	15	45	68,175	2.22% (1)	Class II (HLA-DP)	CRYBG3 p.S316F
8434	Colorectal	74	14	36	37,296	13.9% (5)	Class II (HLA-DRB1)	ARHGEF16 p.R150W
							Class I (HLA-B)	KRAS p.Q61H
0025	Endometrial	36	12	61	26,352	29.5% (18)	Class II (HLA-DRB1)	GFRA2 p.R246H
							Class II (HLA-DRB1)	RHPN2 p.S201C
0894	Breast	115	12	49	67,620	30.6% (15)	Class II (HLA-DRB1)	HNRNPF p.E56K
							Class II (HLA-DRB1)	KDM1A p.D691H
							Class II (HLA-DP)	USP9X p.I1321M
							Class II (HLA-DRB1)	LLGL1 p.E966K
							Class II (HLA-DRB1)	ACO2 p.H719Y
							Class II (HLA-DP)	POLDIP3 p.S400F
8540	Colorectal	68	13	55	48,620	12.7% (7)	Class I (HLA-B)	NUP205 p.R214H
							Class II (HLA-DQ)	PCSK9 p.C477Y
							Class II (HLA-DRB1)	CEP85 p.H549R
5040	Endometrial	150	14	47	98,700	21.3% (10)	Class I (HLA-B)	EMC8 p.T140M
							Class I (HLA-B)	LCK(X1) p.D326G
							Class II (HLA-DRB1)	LCK p.D326G
							Class II (HLA-DRB1)	VARS p.R181C
							Class II (HLA-DP)	RCC1 p.R430C
8202	Colorectal	164	16	56	146,944	8.8% (5)	Class I (HLA-A)	Pending
							Class I (HLA-B)	Pending
							Class II (HLA-DP)	ATP1A1 p.A352T

A total of 5.3×10^5 TCR×HLA×neoantigen permutations were evaluated in nine patients with a mean plexity of 5.8×10^4 per patient. All specimens screened (colorectal n=5, endometrial n=2, breast n=2) yielded at least one neoantigen-reactive TCR. The 63 neoantigen-reactive TCRs (14% of 389 total TCRs screened) identified targeted 22 mutations, including one shared *KRAS* and 21 personal mutations. Of these, 78% (49) were restricted by HLA class II while 22% (14) were restricted by class I. A median reactive hit rate of 13% was achieved per patient (range 2-31%) with an average of three unique neoantigen specificities (range 1-6).

TCRs discovered from hunTR™ platform demonstrate discrete signal to noise ratio

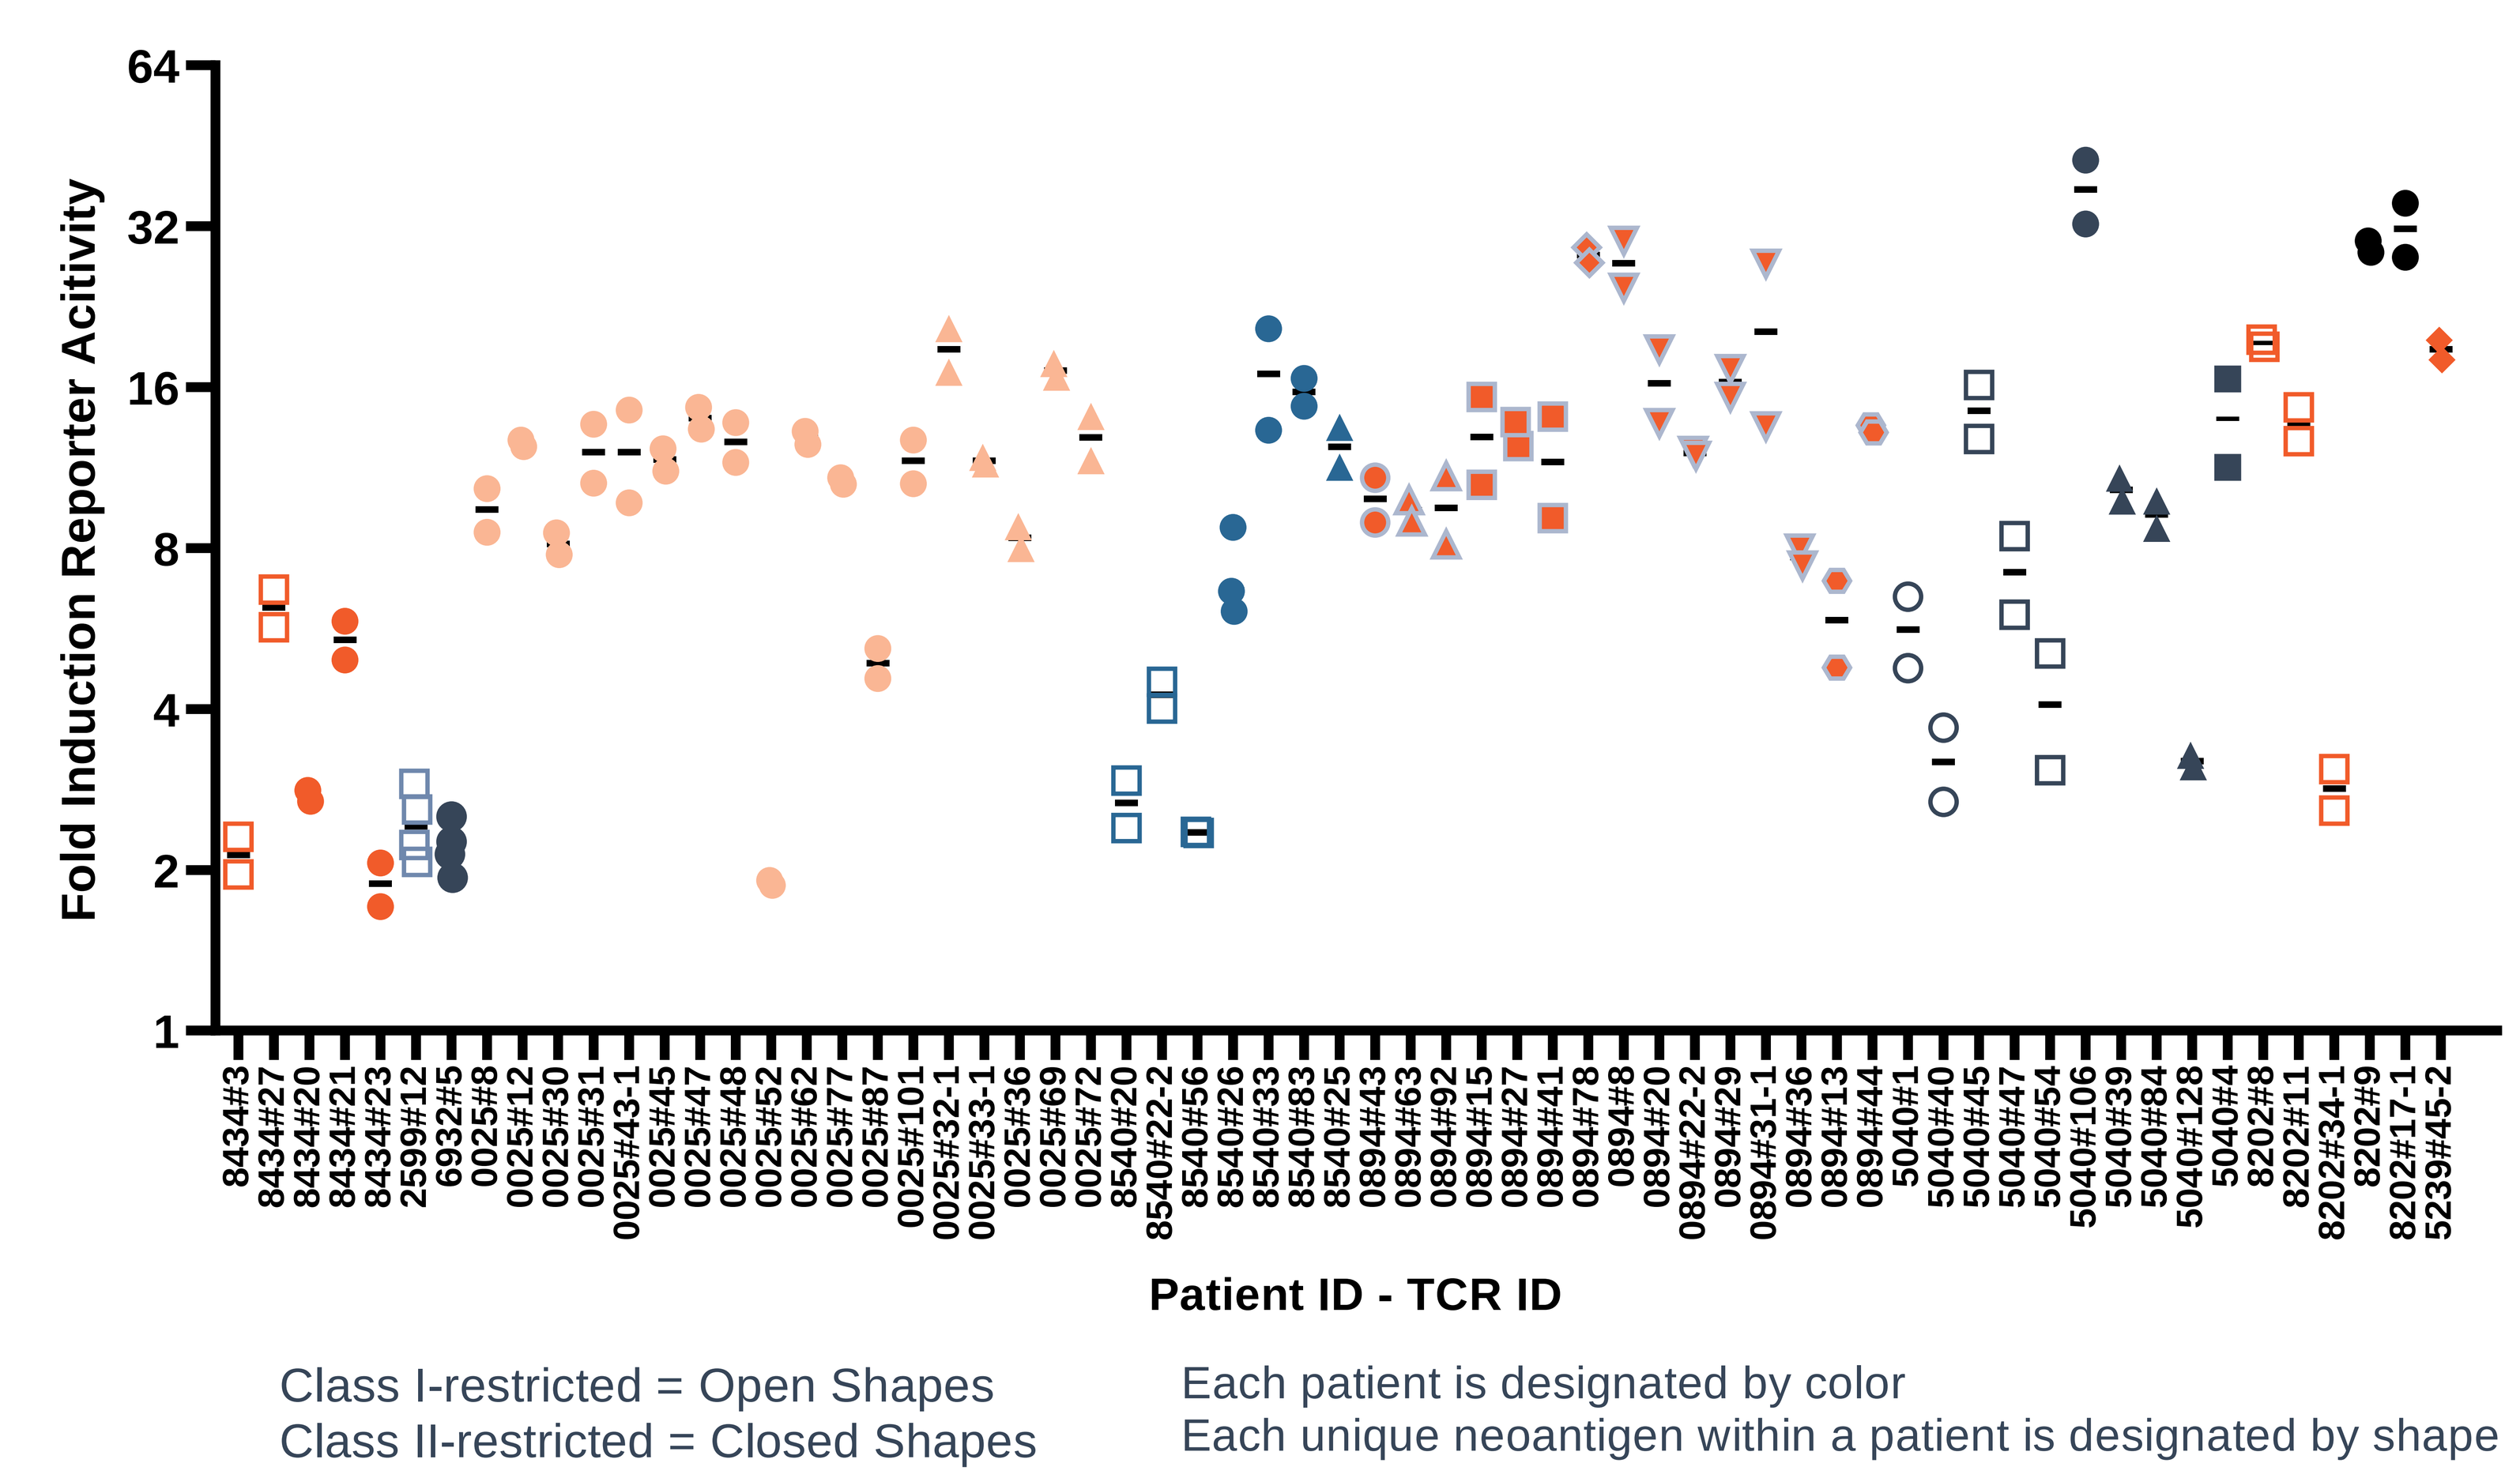
Fold Induction Reporter Acitivity
Patient ID - TCR ID
Class I-restricted = Open Shapes
Class II-restricted = Closed Shapes
Each patient is designated by color
Each unique neoantigen within a patient is designated by shape

Neoantigen specific reporter activity (Fold Induction) is calculated based on negative control cells seeded without any TCR electroporation. All eight patients neo-reactive TCRs were shown with change above 1.5.

hunTR™ platform identified two hotspot KRAS^{Q61H} reactive TCRs in Patient 8434

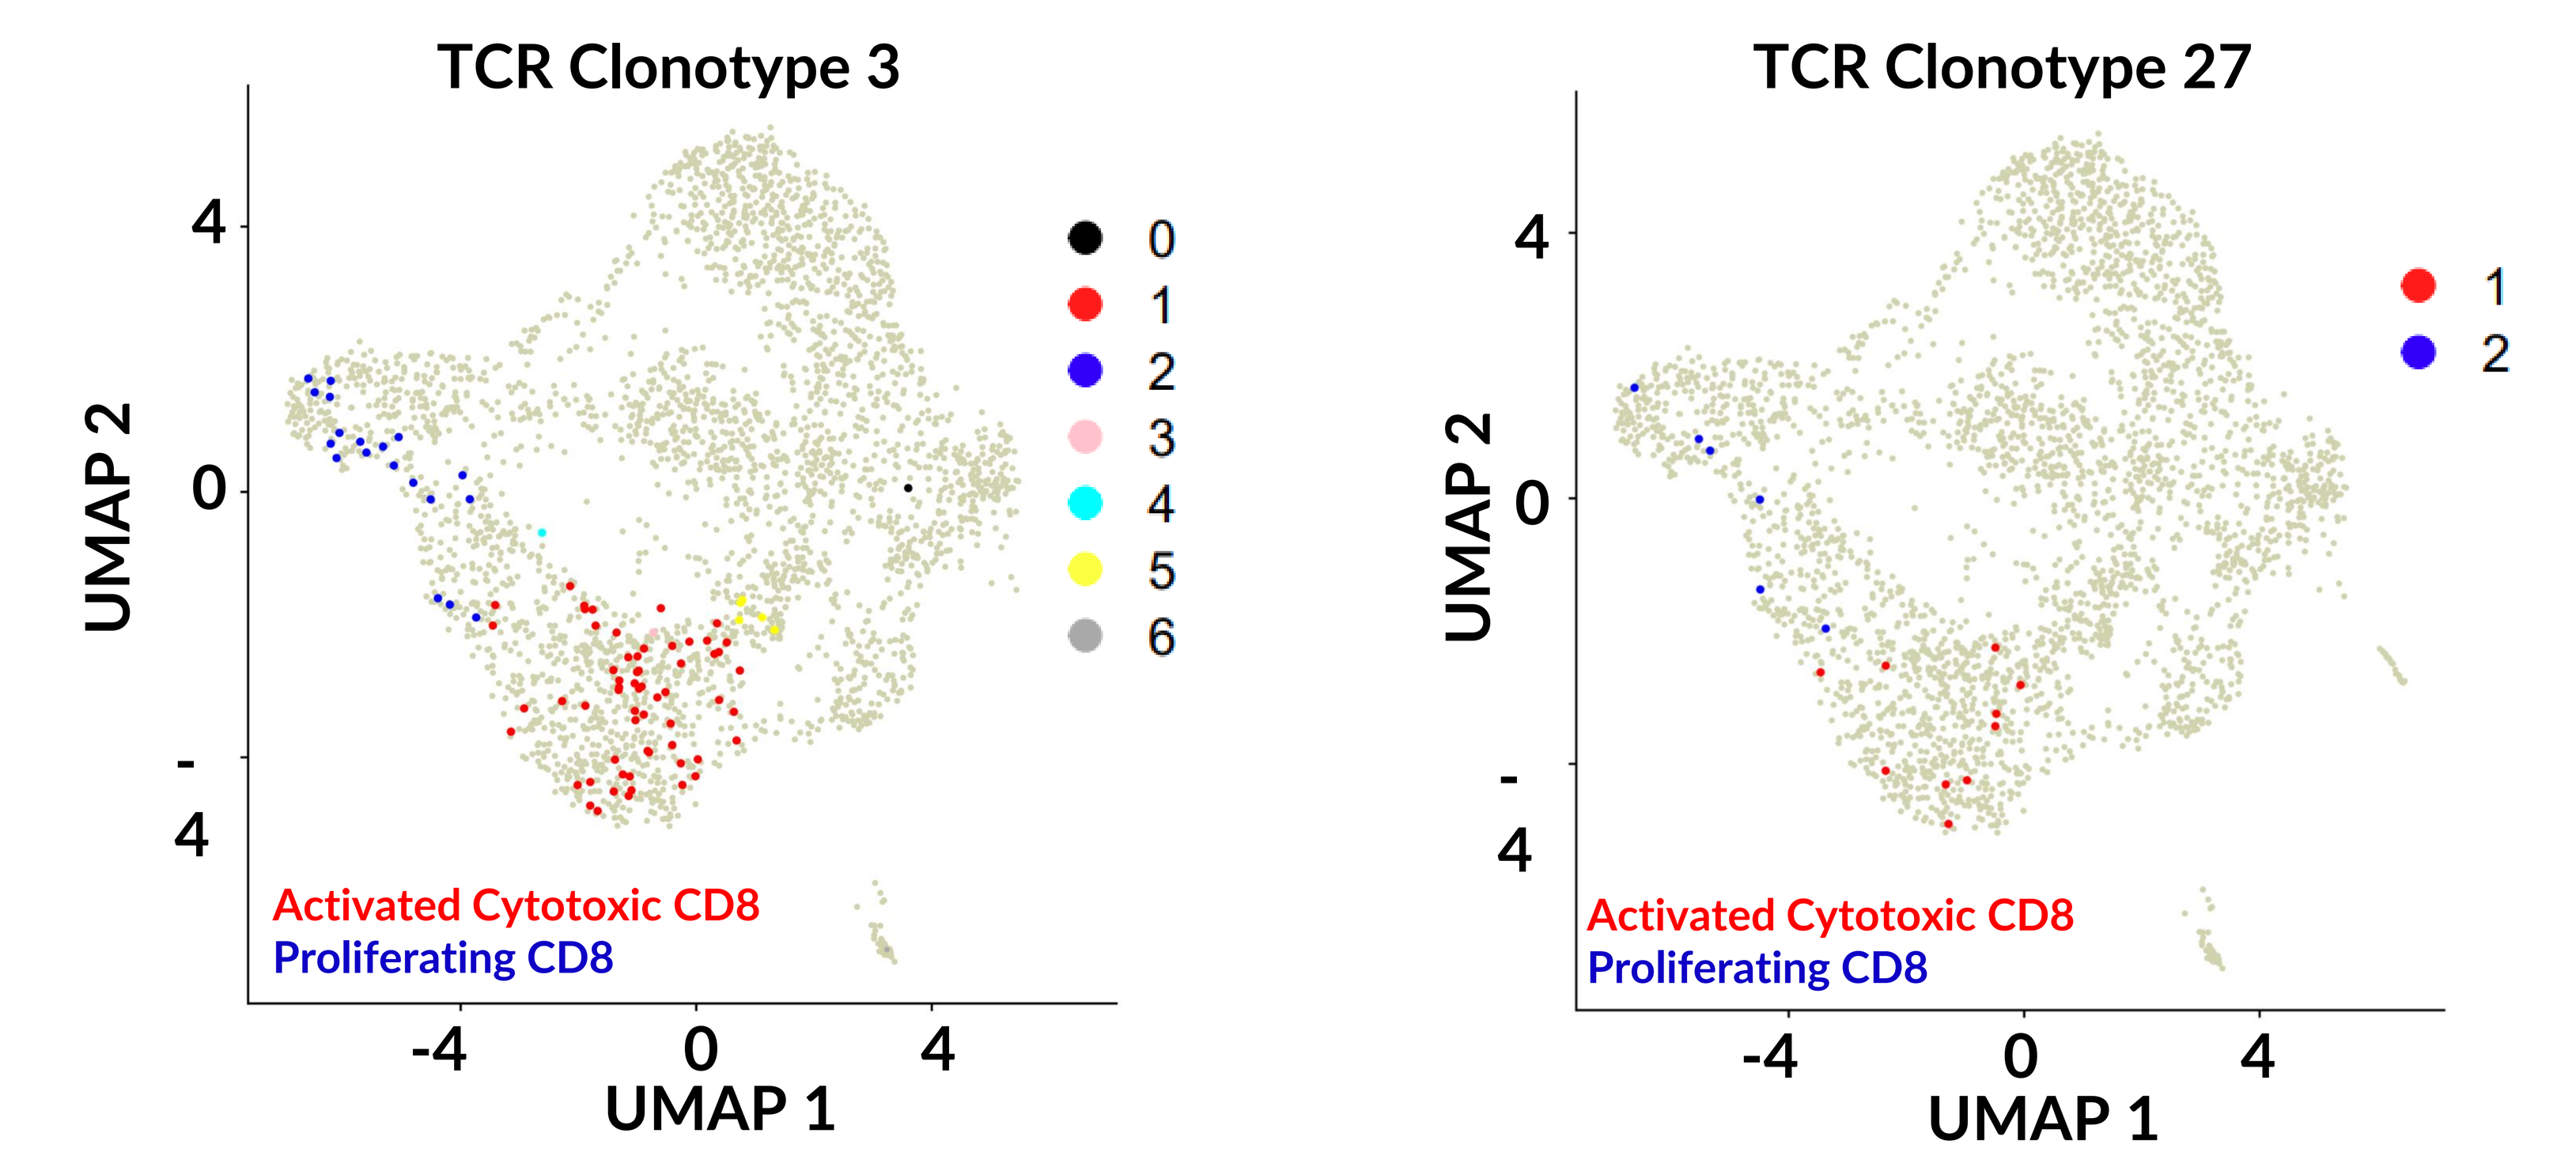
TCR Clonotype 3
TCR Clonotype 27
Activated Cytotoxic CD8
Proliferating CD8
UMAP 1
UMAP 2

TCR clonotype 3 (TCR 3) and TCR clonotype 27 (TCR 27) were found to be reactive to KRAS^{Q61H} with HLA-B. Both TCR Clonotypes 3 and 27 were enriched in clusters 1 and 2. Based on the differential gene expression, cells in cluster 1 are activated CD8 T cells and cells in cluster 2 are proliferating CD8 T cells.

Patient 8434 TCRs displayed differential avidity and specificity to KRAS^{Q61H}

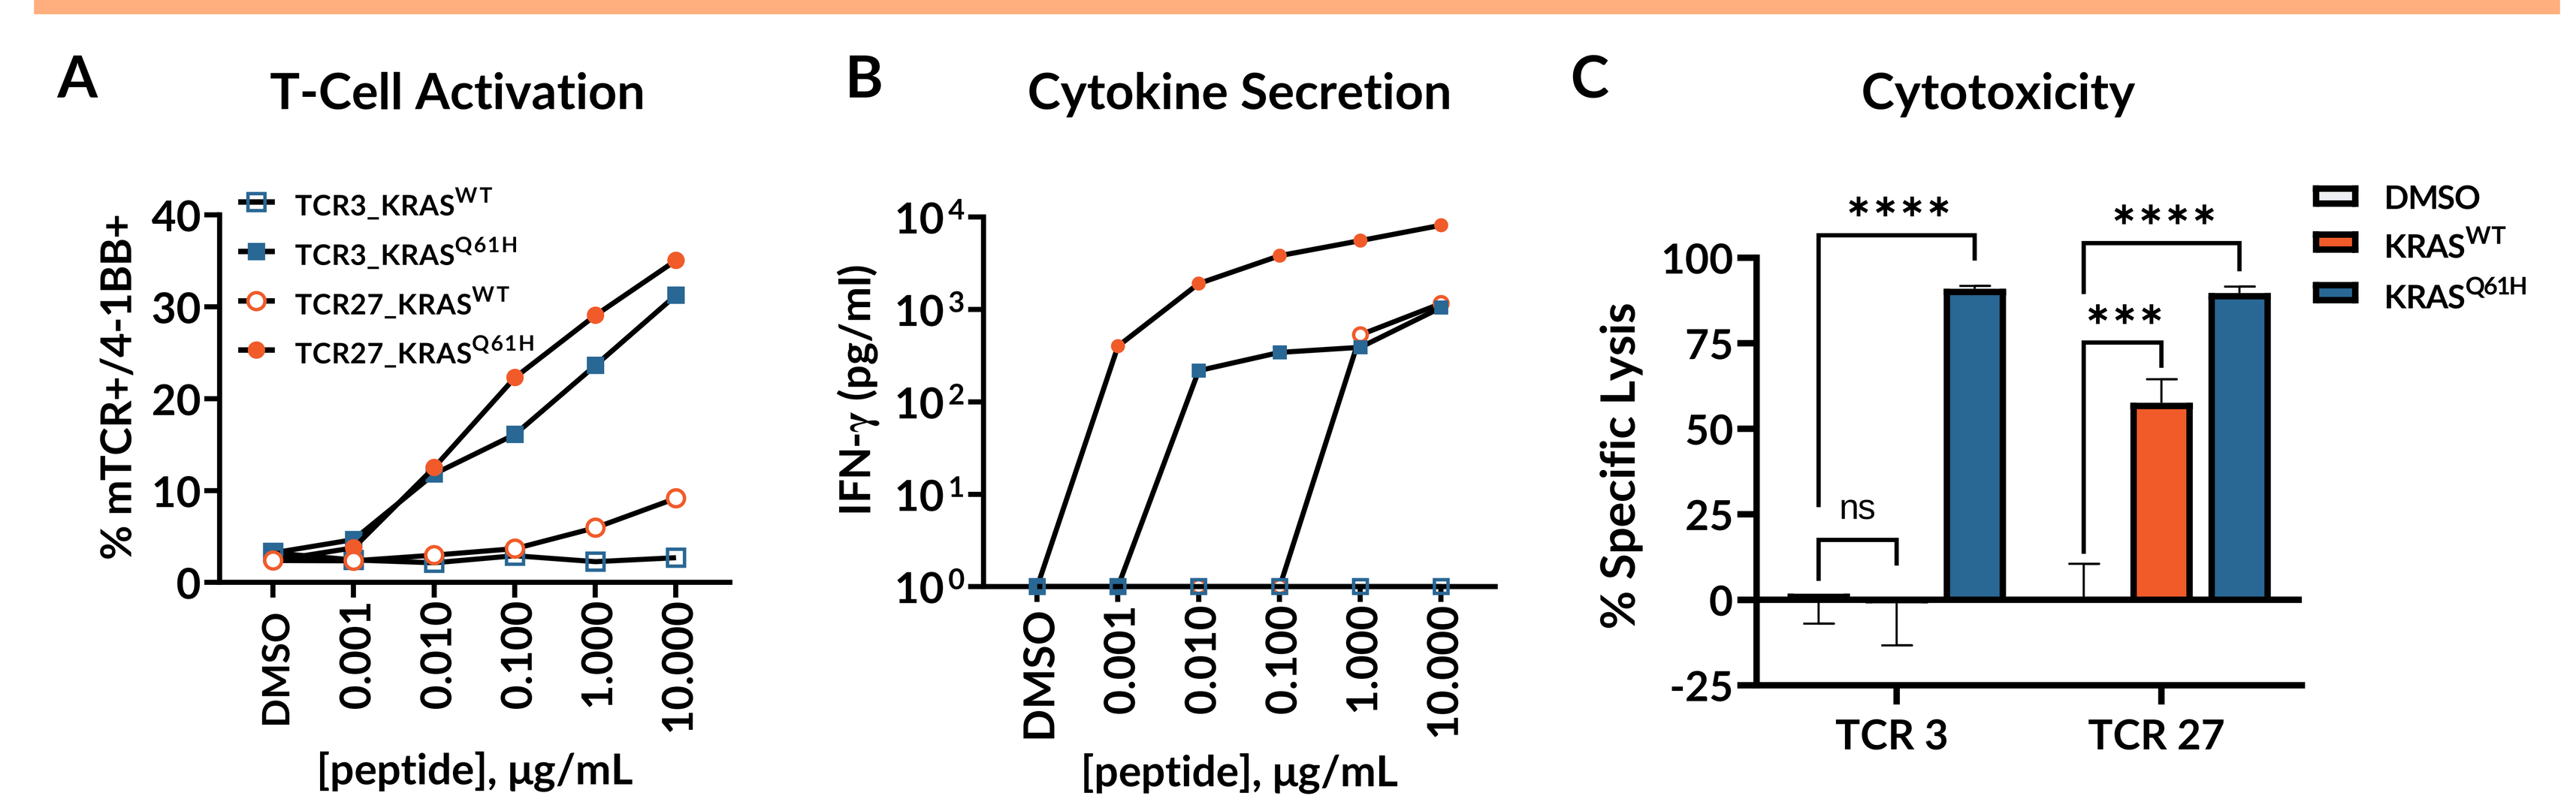
A T-Cell Activation
TCR3_KRASWT
TCR3_KRASQ61H
TCR27_KRASWT
TCR27_KRASQ61H
% mTCR+/4-1BB+
[peptide], µg/mL
B Cytokine Secretion
IFN-γ (pg/ml)
[peptide], µg/mL
C Cytotoxicity
DMSO
KRASWT
KRASQ61H
% Specific Lysis
TCR 3
TCR 27

Primary human TCR-T cells were co-cultured with antigen presenting cells pulsed with either wild or mutant KRAS peptides. T-cell activation was measured by (A) surface 4-1BB expression and (B) IFN-γ secretion. (C) Cytolytic activity against neoantigen presenting tumor cells was measured in a tumor cell line after cocultured with TCR-T cells.

Targeted *KRAS* mutation screening yields reactive TCRs from 2/5 CRC Patients

KRAS Patient Screening Results				
Patient ID	Cancer Diagnosis	KRAS Mutation	HLA Restriction	Mutation Specificity
1036	Colorectal	KRAS p.G12V	N/A	N/A
3080	Colorectal	KRAS p.G12D	N/A	N/A
9921	Colorectal	KRAS p.G12V	N/A	N/A
9976	Colorectal	KRAS p.G12V	DRB1*07:01	KRAS p.G12V
7014	Colorectal	KRAS p.G12V	DRB1*07:01	KRAS p.G12V

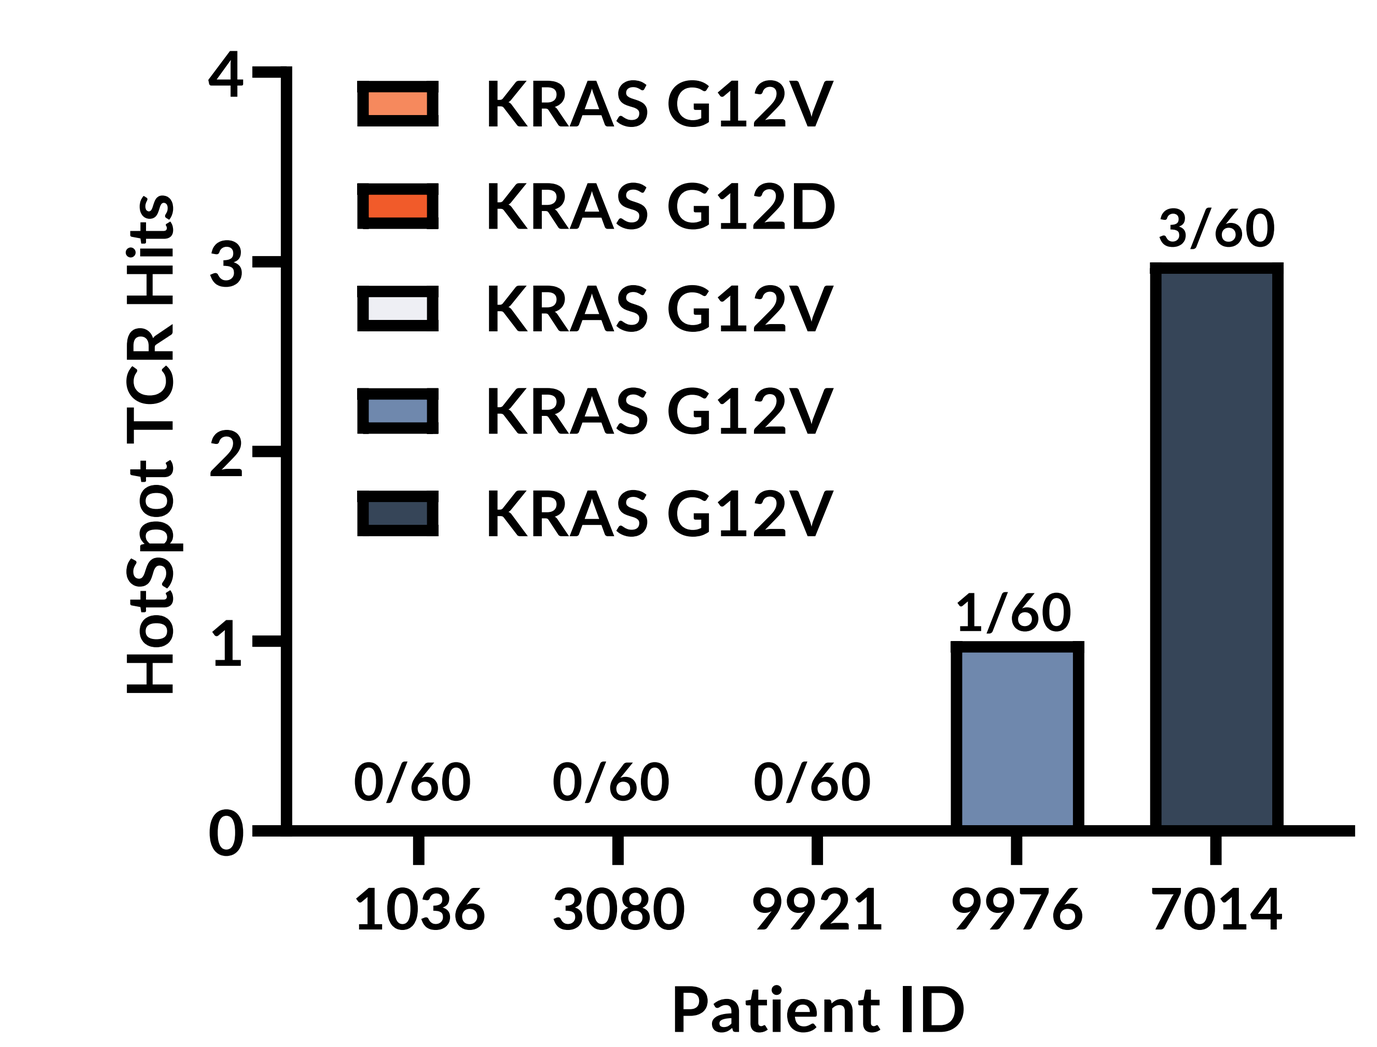
KRAS G12V
KRAS G12D
KRAS G12V
KRAS G12V
KRAS G12V
HotSpot TCR Hits
0/60
0/60
0/60
1/60
3/60
1036
3080
9921
9976
7014
Patient ID

Four KRAS mutation reactive TCRs found through targeted hunting against *KRAS* mutations in five colorectal cancer (CRC) patients (40% success rate; Results Table [left] and Figure [right]). They were all against KRAS G12V and restricted to HLA DRB1*07:01. Hot-spot mutation targeted screening method has the potential to drive rapid hotspot mutation-specific TCR discovery. Values on top of bar graph indicate *KRAS* mutation-reactive TCRs discovered over TCRs screened.

hunTR™ is designed to expand patient population eligible for TCR-T cell therapy

hunTR™ is a hyperplex screening platform that identifies neoantigen-reactive TCRs. hunTR™ allows for the expansion of Alaunos' hotspot mutation-targeted TCR library, increasing the addressable population of solid tumor patients (with matching hotspot mutation and HLA allele) eligible for TCR-T cell treatment. In addition, hunTR™ is applicable for personalized TCR-T therapy such that most solid tumor patients could be eligible for mutation-targeted cell therapy.